Illimis therapeutics

KAIST

Anti-inflammatory clearance of amyloid beta by a chimeric Gas6 fusion protein

Min Soo Kang[1], Hyuncheol Jung[1,2], Se Young Lee[2], Sukhee Cho[1], Ah-Young Shin[1], Jaekeun Lee[1], Sanghoon Park[1], Won-Suk Chung[2], Chan Hyuk Kim[2]

[1]Illimis Therapeutics, Inc., Seoul, Republic of Korea, [2]Department of Biological Sciences, Korea Advanced Institute of Science and Technology (KAIST), Daejeon, Republic of Korea

INTRODUCTION

Unmet needs in Aβ-targeting immunotherapy

- Aβ immunotherapy is one of the most promising approaches in treating Alzheimer's disease (AD).
- Although **Aβ-targeting antibodies can substantially reduce Aβ burden in the brain** via Fc receptor-mediated phagocytosis, they **concurrently release pro-inflammatory cytokines and chemokines** which lead to marginal effects on cognitive function and safety concerns.
- **Amyloid-associated imaging abnormalities (ARIA), which are found in various types of brain edema and microhemorrhage, are frequently observed in patients following Aβ-targeting antibody therapy**. These anomalies are intimately linked to immune responses and inflammation, and there have been instances of individuals receiving Aβ-antibody experiencing ventricular enlargement and a sign of white and gray matter shrinkage, which are closely connected to ARIA.
- Innovations for removing Aβ without inflammatory responses are in demand.

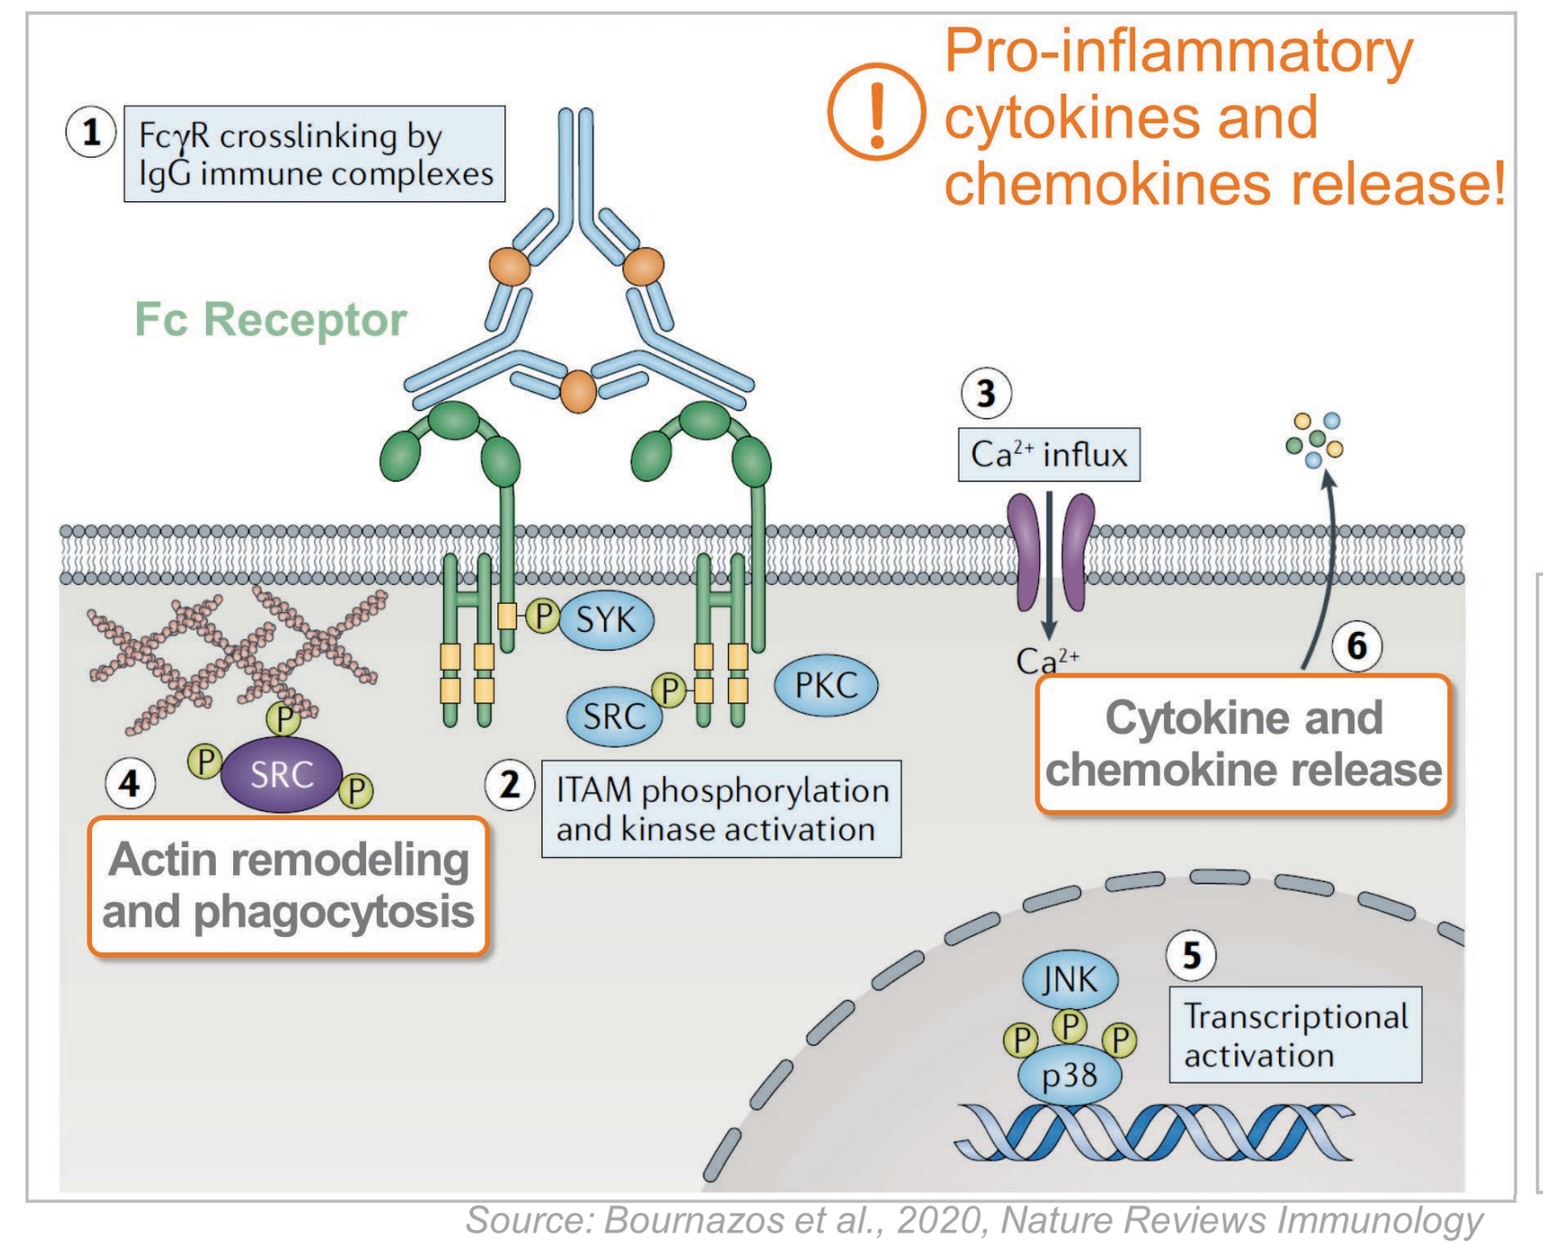
FcγR crosslinking by IgG immune complexes
Pro-inflammatory cytokines and chemokines release!
Fc Receptor
Ca²⁺ influx
SYK
SRC
PKC
ITAM phosphorylation and kinase activation
Actin remodeling and phagocytosis
Cytokine and chemokine release
JNK
p38
Transcriptional activation

Source: Bournazos et al., 2020, Nature Reviews Immunology

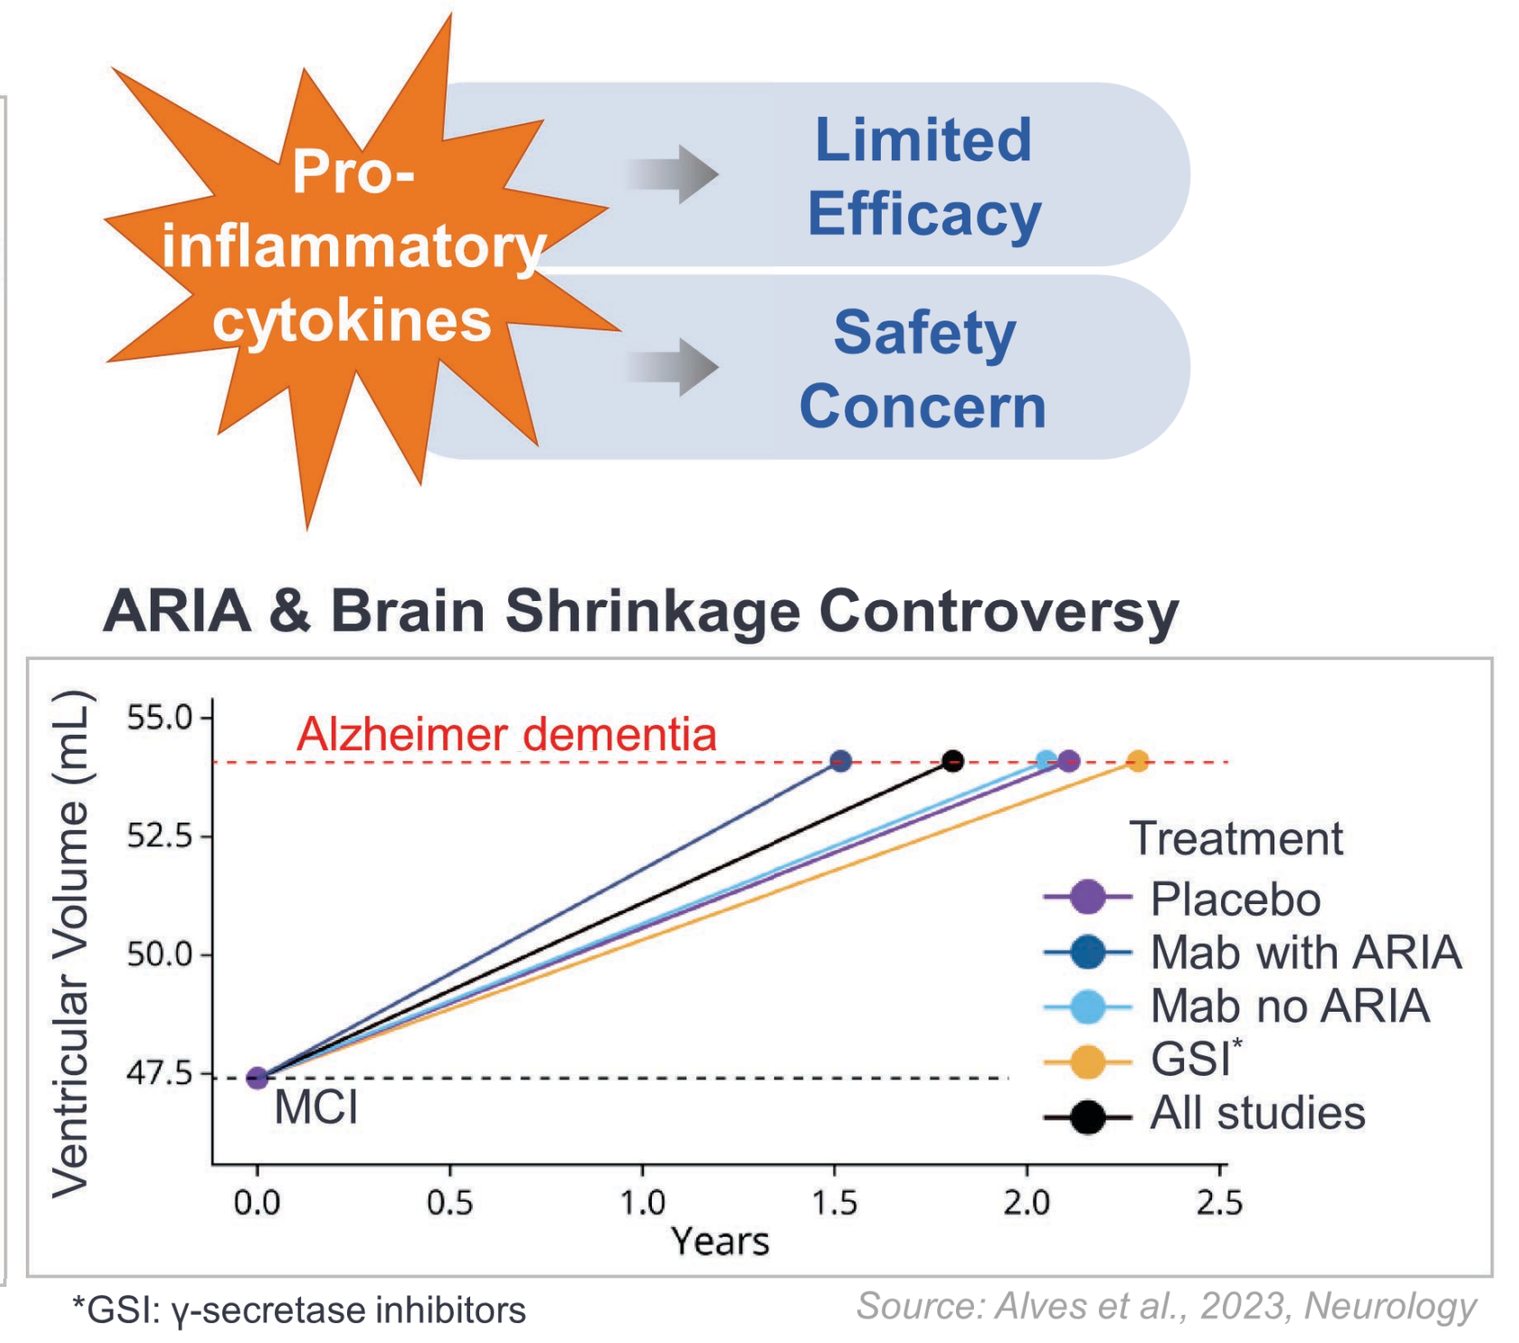
Pro-inflammatory cytokines
Limited Efficacy
Safety Concern
ARIA & Brain Shrinkage Controversy
Alzheimer dementia
MCI
Ventricular Volume (mL)
Years
Treatment
Placebo
Mab with ARIA
Mab no ARIA
GSI*
All studies

*GSI: γ-secretase inhibitors

Source: Alves et al., 2023, Neurology

Efferocytosis: TAM-mediated anti-inflammatory phagocytosis

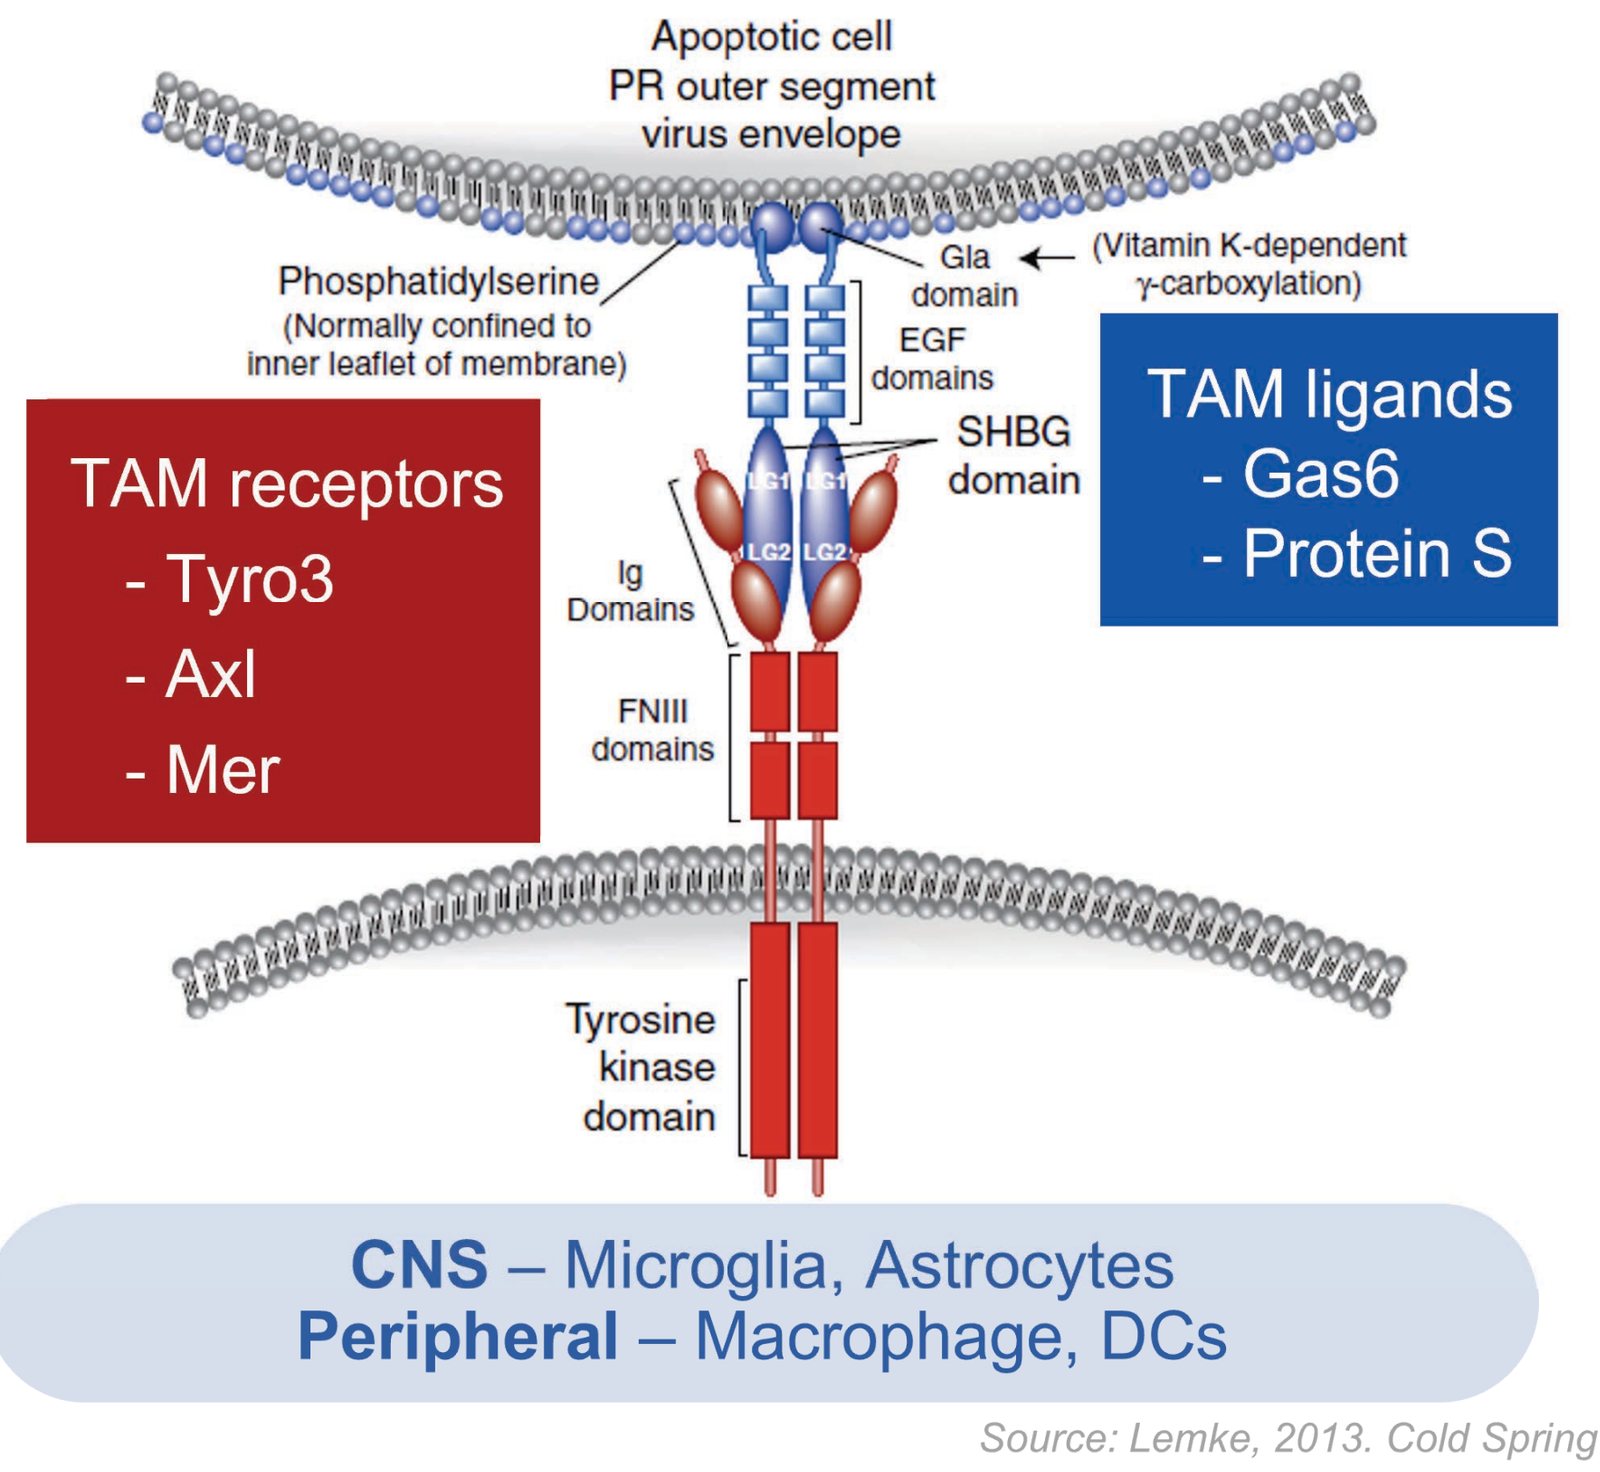
Apoptotic cell
PR outer segment
virus envelope
Phosphatidylserine (Normally confined to inner leaflet of membrane)
Gla domain
(Vitamin K-dependent γ-carboxylation)
EGF domains
SHBG domain
TAM receptors
- Tyro3
- Axl
- Mer
TAM ligands
- Gas6
- Protein S
Ig Domains
FNIII domains
Tyrosine kinase domain
CNS – Microglia, Astrocytes
Peripheral – Macrophage, DCs

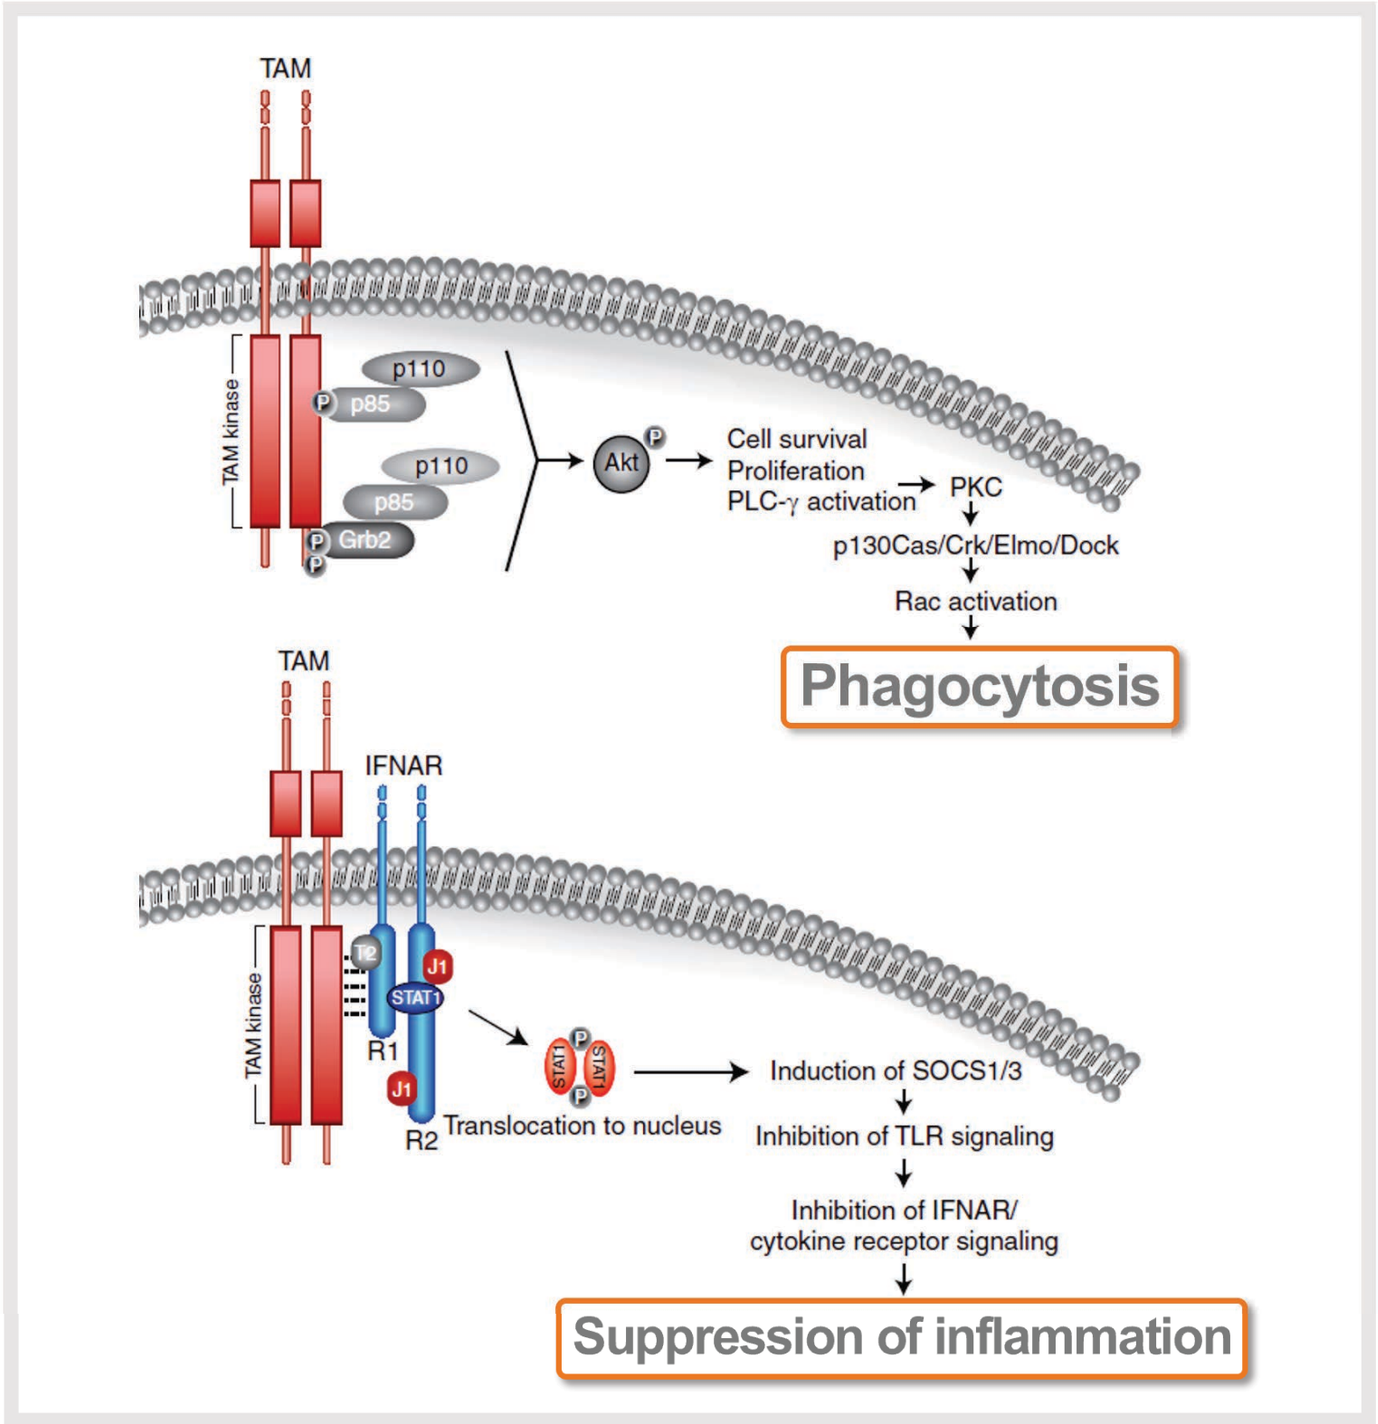
TAM
Cell survival
Proliferation
PLC-γ activation
PKC
p130Cas/Crk/Elmo/Dock
Rac activation
Phagocytosis
IFNAR
Translocation to nucleus
Induction of SOCS1/3
Inhibition of TLR signaling
Inhibition of IFNAR/ cytokine receptor signaling
Suppression of inflammation

Source: Lemke, 2013. Cold Spring Harb Perspect Biol

- For cellular homeostasis maintenance, phagocytes remove apoptotic cells and unnecessary proteins via **efferocytosis which induces phagocytosis accompanied with suppression of inflammation**.
- **The Tyro3, Axl, and Mer (TAM) receptors and their ligand Gas6 induce efferocytosis**. The ligand Gas6 causes efferocytosis by connecting the TAM receptors and the target, such as phosphatidylserine of apoptotic cells, on a variety of cells that express TAM receptors.

STRATEGY

Illimis' Platform Technology

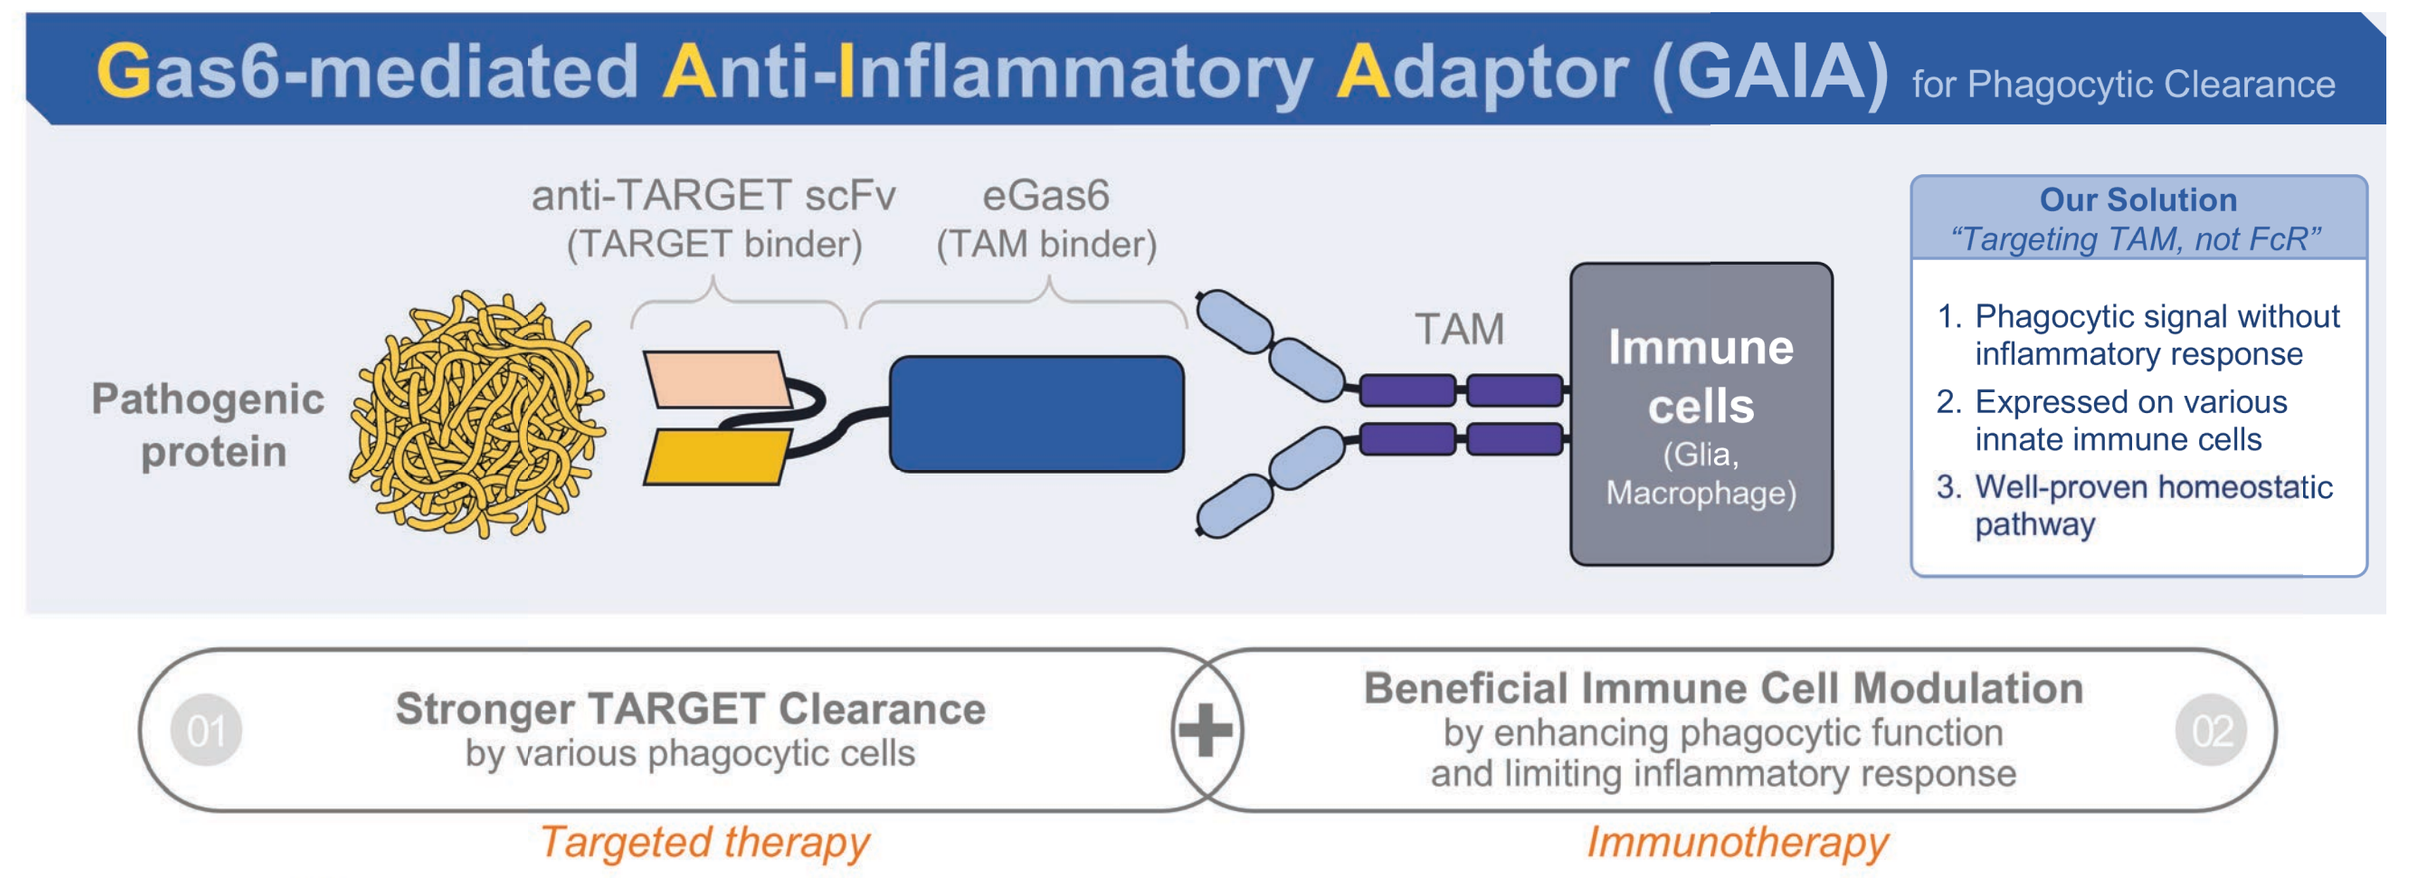
Gas6-mediated Anti-Inflammatory Adaptor (GAIA) for Phagocytic Clearance
anti-TARGET scFv (TARGET binder)
eGas6 (TAM binder)
Pathogenic protein
TAM
Immune cells (Glia, Macrophage)
Our Solution
"Targeting TAM, not FcR"
1. Phagocytic signal without inflammatory response
2. Expressed on various innate immune cells
3. Well-proven homeostatic pathway
01 Stronger TARGET Clearance by various phagocytic cells
Targeted therapy
Beneficial Immune Cell Modulation by enhancing phagocytic function and limiting inflammatory response 02
Immunotherapy

"Clearing pathogenic protein without unwanted neuroinflammation"

- By using minimal active domain of Gas6, we developed a novel fusion protein platform "GAIA" which enables the target-directed phagocytosis without unwanted neuroinflammation.
- GAIA consists of two parts: **1) TARGET binder** and **2) TAM binder**
 Once GAIA binds both the pathogenic protein (e.g. Aβ) and TAM receptors on glial cells, it induces efferocytosis enabling efficient removal of the target proteins and limiting inflammatory responses simultaneously.
- As the **target binder is interchangeable, target indication can be easily expanded from neurodegenerative diseases to various immune-related diseases.**
- We constructed αAβ-Gas6 as a prototype in AD to confirm proof-of-concept of GAIA.

RESULTS

αAβ-Gas6

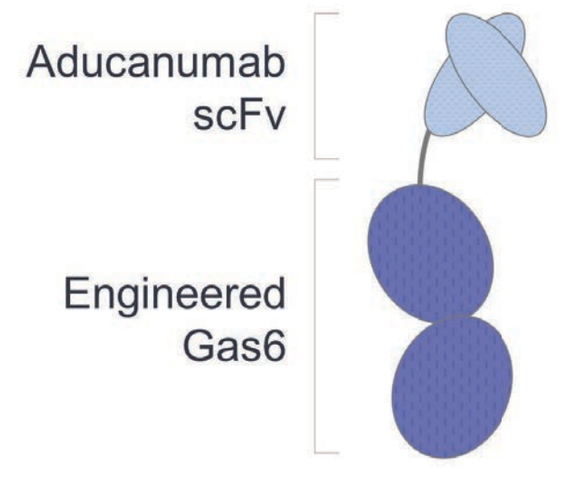
Aducanumab scFv
Engineered Gas6

Phagocytosis

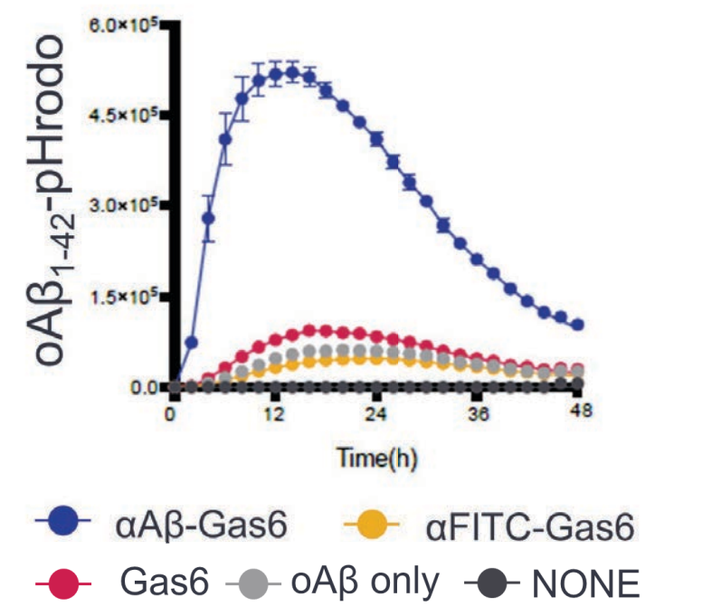
oAβ1-42-pHrodo
Time(h)
αAβ-Gas6
αFITC-Gas6
Gas6
oAβ only
NONE

Anti-inflammatory effects

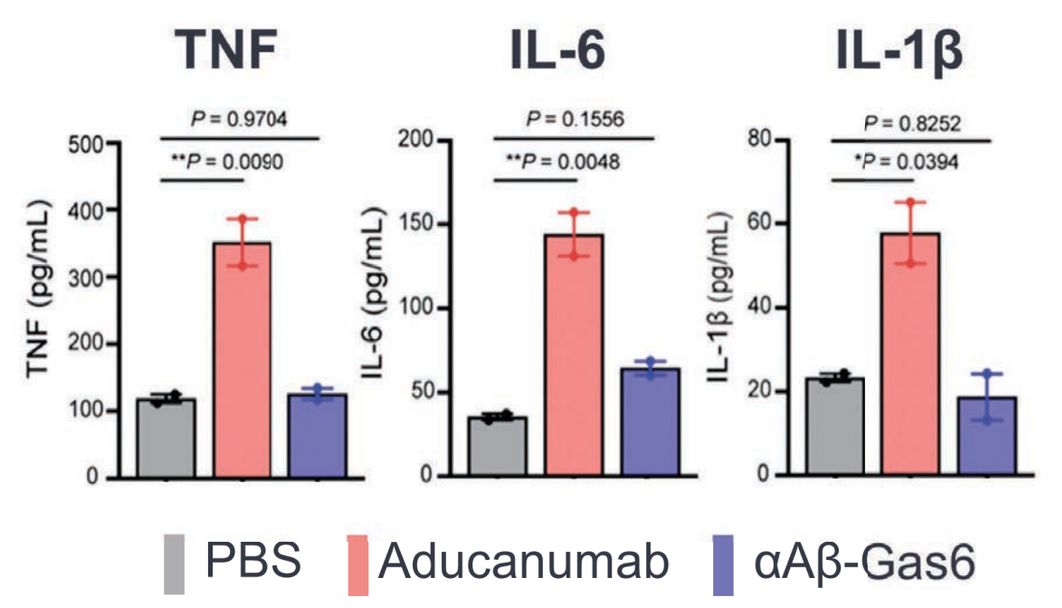
TNF
IL-6
IL-1β
P = 0.9704
**P = 0.0090
P = 0.1556
**P = 0.0048
P = 0.8252
*P = 0.0394
TNF (pg/mL)
IL-6 (pg/mL)
IL-1β (pg/mL)
PBS
Aducanumab
αAβ-Gas6

- **Phagocytosis** - to confirm the target-selective uptake, we used oligomeric Aβ conjugated with pHrodo (oAβ-pHrodo) serving as a reporter of phagocytic internalization. Live-cell imaging analysis showed that only αAβ-Gas6-treated cells exhibit a significantly increased phagocytosis of oligomeric Aβ compared to controls.
- **Anti-inflammatory effect** - Quantitative bar graphs show the protein levels of pro-inflammatory cytokines (TNF, IL-6, and IL-1β) were low in αAβ-Gas6 treated groups confirming phagocytic uptake of oligomeric Aβ without inflammation.

Reduced Neurotoxicity

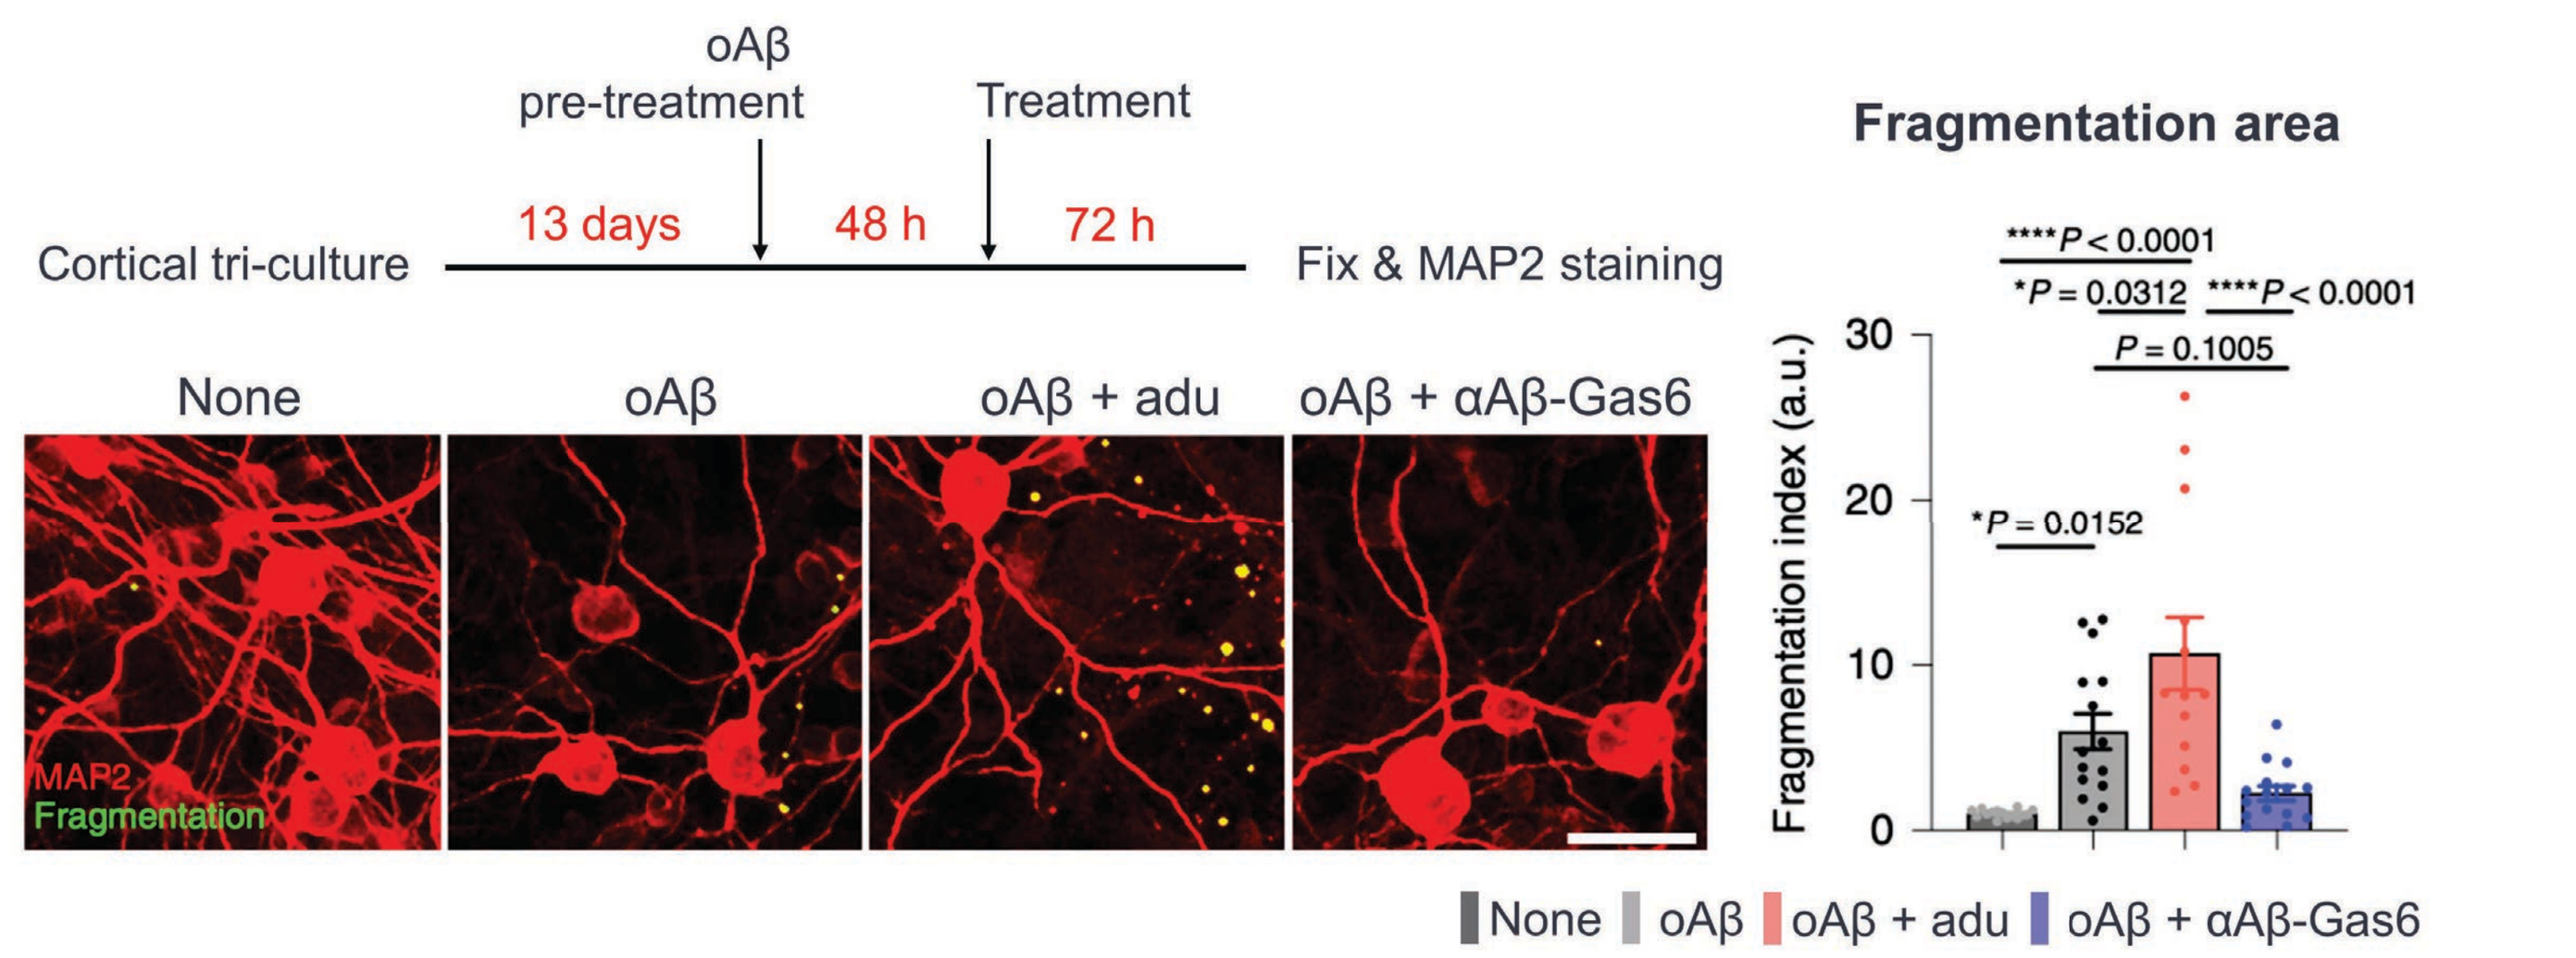
oAβ pre-treatment
Treatment
Cortical tri-culture
13 days
48 h
72 h
Fix & MAP2 staining
None
oAβ
oAβ + adu
oAβ + αAβ-Gas6
MAP2
Fragmentation
Fragmentation area
****P < 0.0001
*P = 0.0312
****P < 0.0001
P = 0.1005
*P = 0.0152
Fragmentation index (a.u.)
None
oAβ
oAβ + adu
oAβ + αAβ-Gas6

- **Reduced neurotoxicity -** Aducanumab significantly exacerbated oAβ-induced neuronal toxicity, resulting in increased neuronal fragmentation. We found that αAβ-Gas6 treatment was able to largely rescue oAβ-induced neuronal fragmentation to levels similar to the oAβ-nontreated control group.

RESULTS

Experimental scheme & Plaque lowering effect

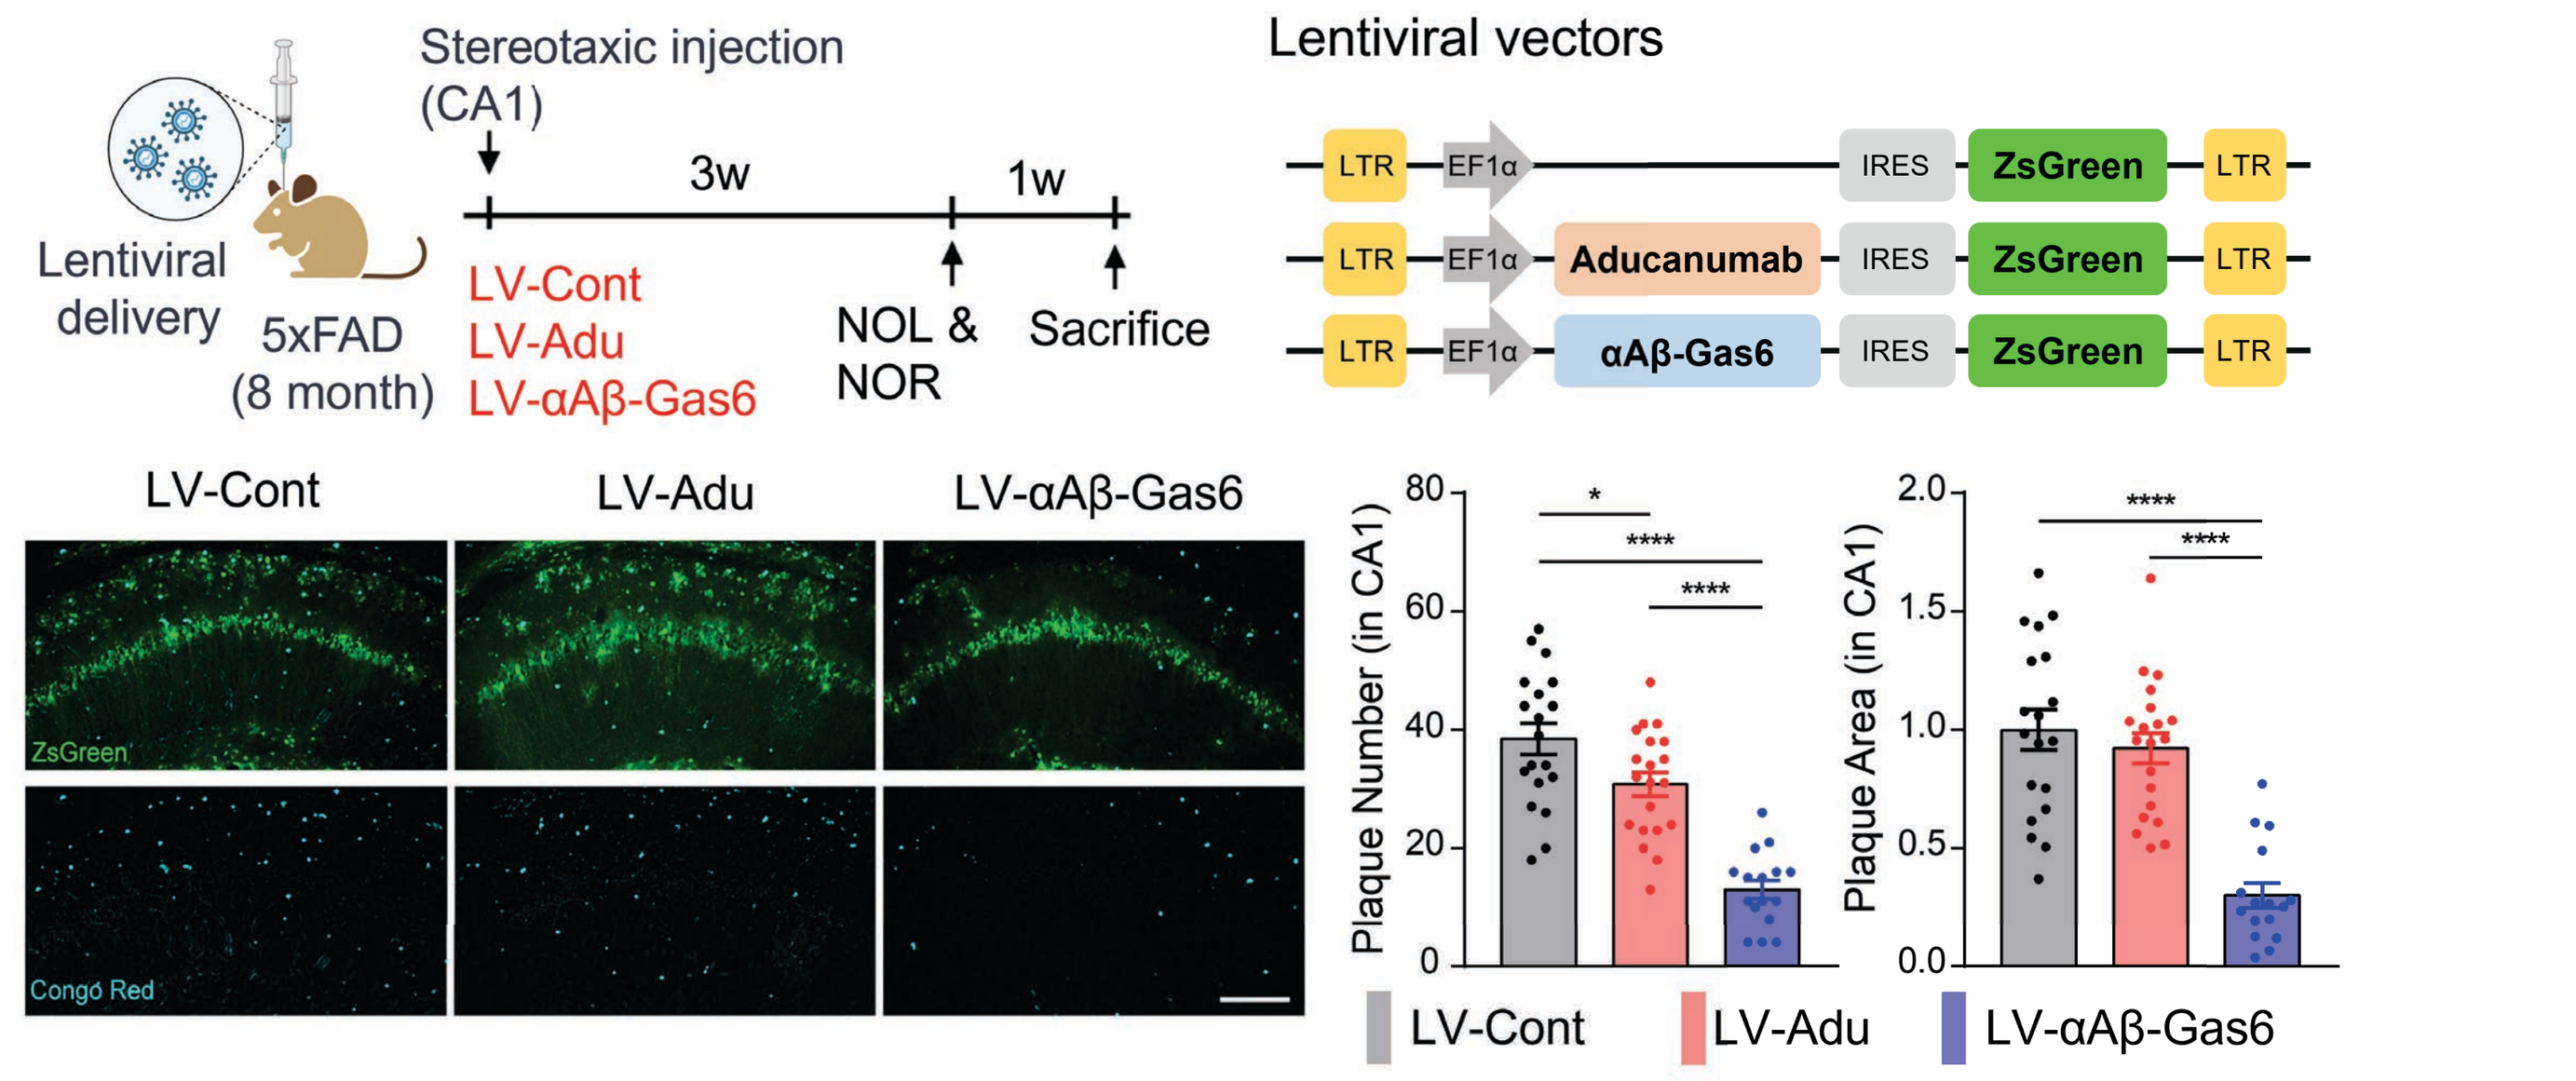
Lentiviral delivery
5xFAD (8 month)
Stereotaxic injection (CA1)
3w
1w
LV-Cont
LV-Adu
LV-αAβ-Gas6
NOL & NOR
Sacrifice
Lentiviral vectors
LTR
EF1α
IRES
ZsGreen
Aducanumab
αAβ-Gas6
LV-Cont
LV-Adu
LV-αAβ-Gas6
ZsGreen
Congo Red
Plaque Number (in CA1)
Plaque Area (in CA1)
LV-Cont
LV-Adu
LV-αAβ-Gas6

- **Plaque lowering effect** - Both the number and total area of Aβ plaques were significantly decreased in the 5xFAD CA1 injected with LV-αAβ-Gas6, but the decrease was minimal with LV-Aducanumab treatment.

Reduced synapse elimination

Improved cognitive function

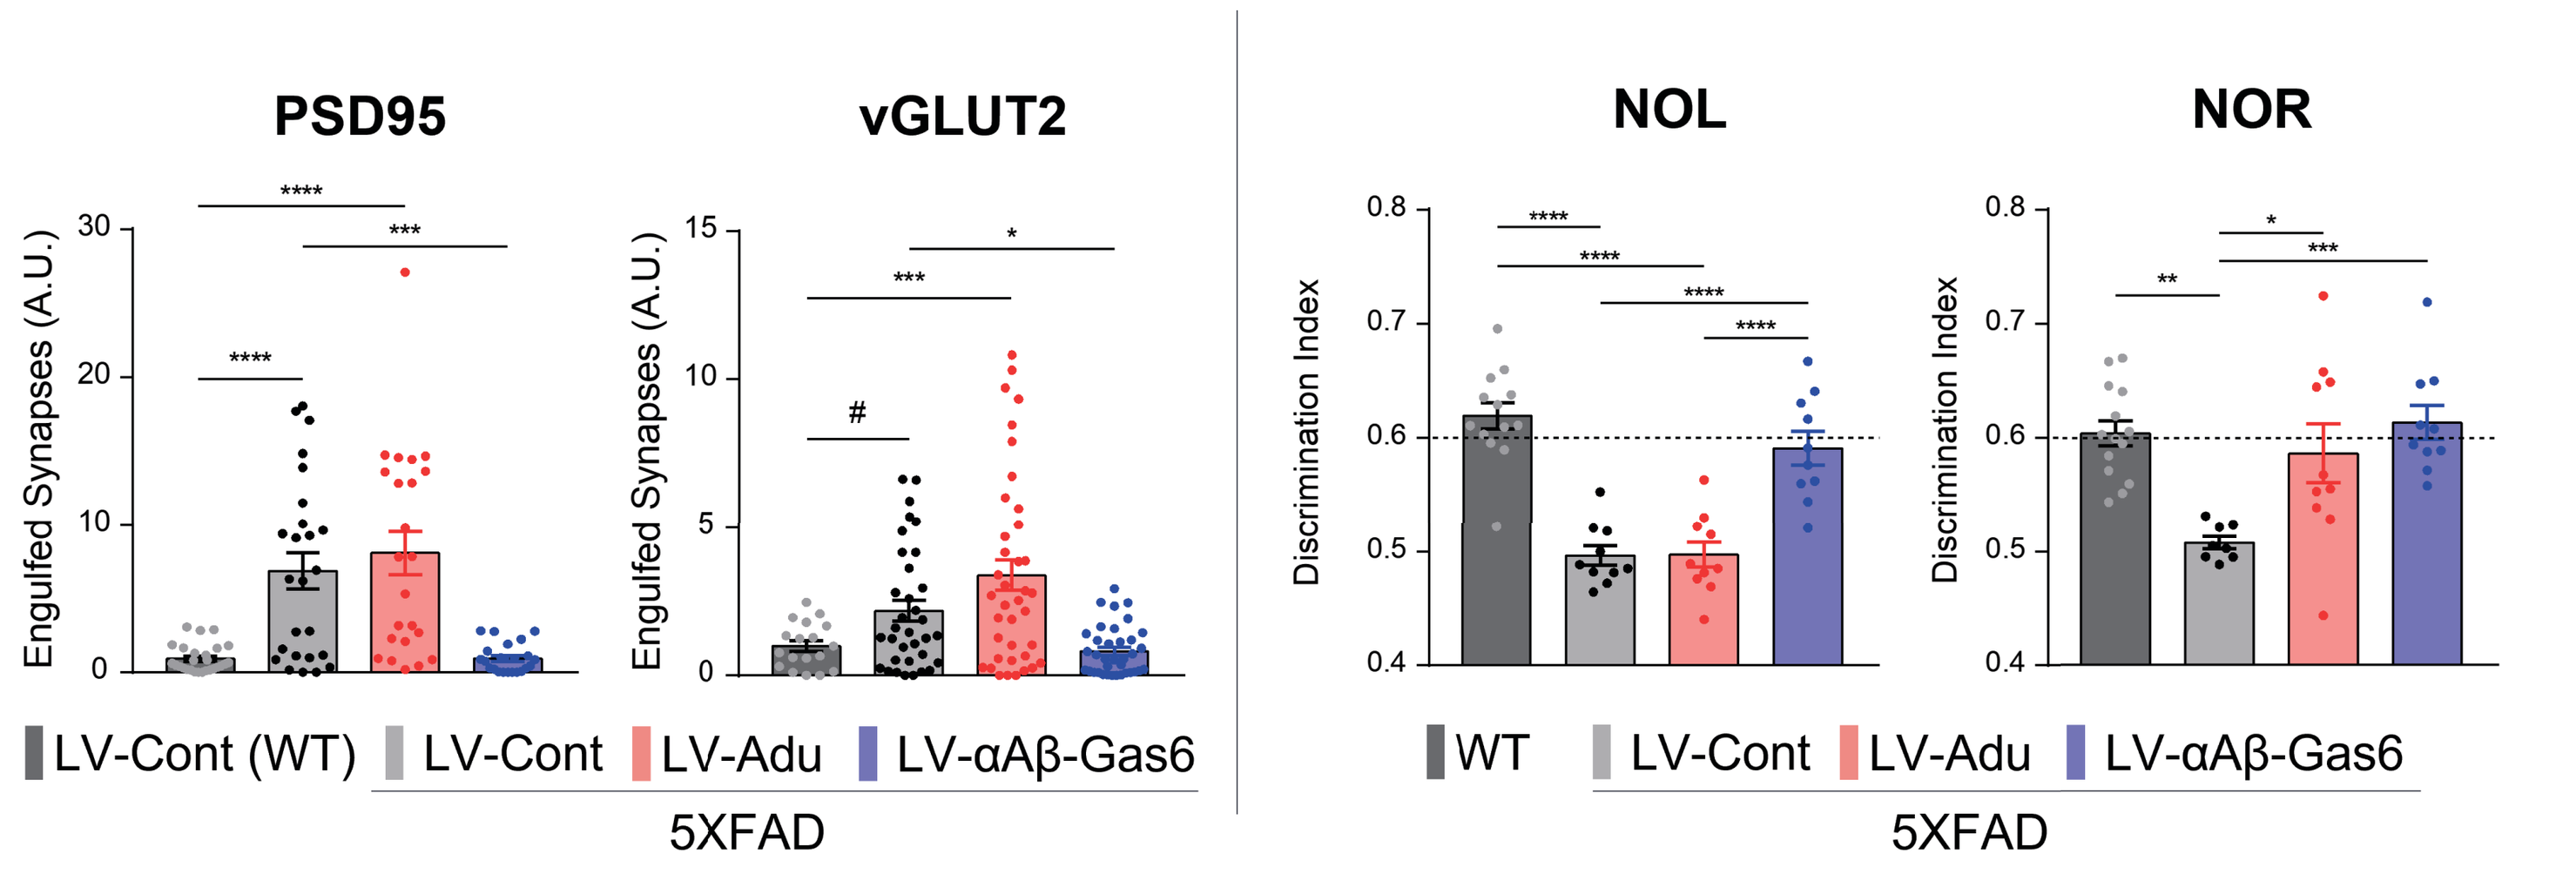
PSD95
vGLUT2
Engulfed Synapses (A.U.)
LV-Cont (WT)
LV-Cont
LV-Adu
LV-αAβ-Gas6
5XFAD
NOL
NOR
Discrimination Index
WT
LV-Cont
LV-Adu
LV-αAβ-Gas6
5XFAD

- **Reduced Synapse elimination** - LV-Adu sustained increased synaptic engulfment levels whereas LV-αAβ-Gas6 injection significantly reduced excessive synapse elimination to levels comparable to non-AD WT.
 This suggests αAβ-Gas6 exhibit therapeutic effects against AD not only by eliminating Aβ plaques but also by preventing excessive synapse elimination through reduction of NPAM activation.
- **Improved cognitive function** - LV-αAβ-Gas6 show normal discrimination index similar to WT in novel object location (NOL) and novel object recognition (NOR) tests, suggesting their cognitive functions were rescued.

Reduced & minimal ARIA

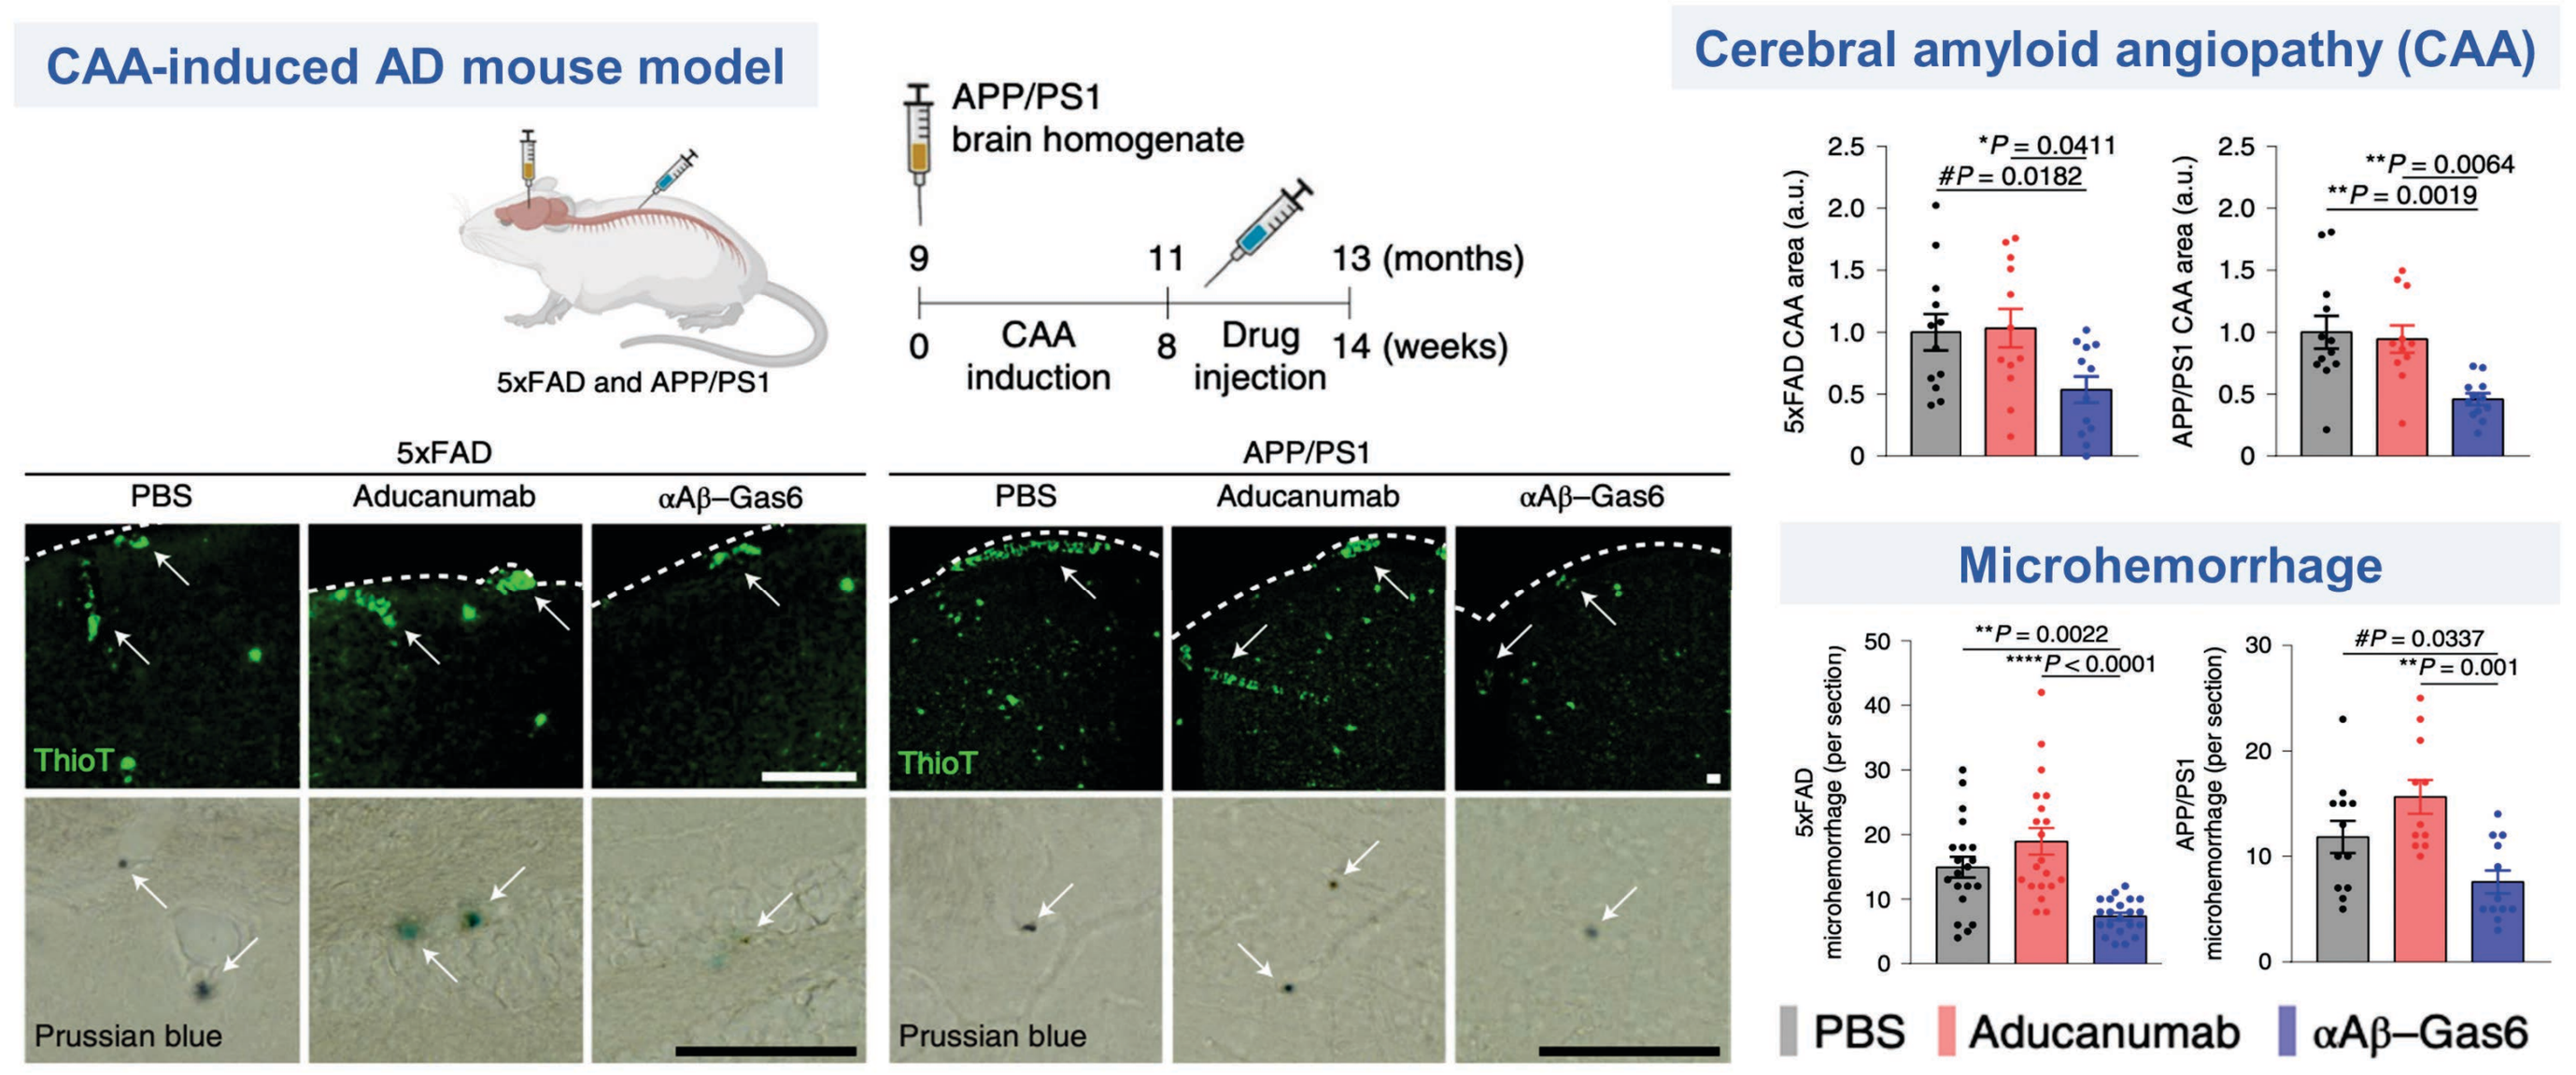
CAA-induced AD mouse model
5xFAD and APP/PS1
APP/PS1 brain homogenate
9
11
13 (months)
0
CAA induction
8
Drug injection
14 (weeks)
5xFAD
APP/PS1
PBS
Aducanumab
αAβ–Gas6
ThioT
Prussian blue
Cerebral amyloid angiopathy (CAA)
*P = 0.0411
#P = 0.0182
**P = 0.0064
**P = 0.0019
5xFAD CAA area (a.u.)
APP/PS1 CAA area (a.u.)
Microhemorrhage
**P = 0.0022
****P < 0.0001
#P = 0.0337
**P = 0.001
5xFAD microhemorrhage (per section)
APP/PS1 microhemorrhage (per section)
PBS
Aducanumab
αAβ–Gas6

- **Reduced CAA** - In the CAA-induced AD mouse model, αAβ-Gas6-injected group showed a significant decrease in both CAA and microhemorrhage, whereas the Aducanumab-injected group showed an increasing tendency of microhemorrhage. The results suggest that αAβ-Gas6 eliminates Aβ plaques without inducing CAA-related side effects of conventional Aβ antibody therapy, such as ARIA.

Anti-inflammatory effects – scRNA-seq

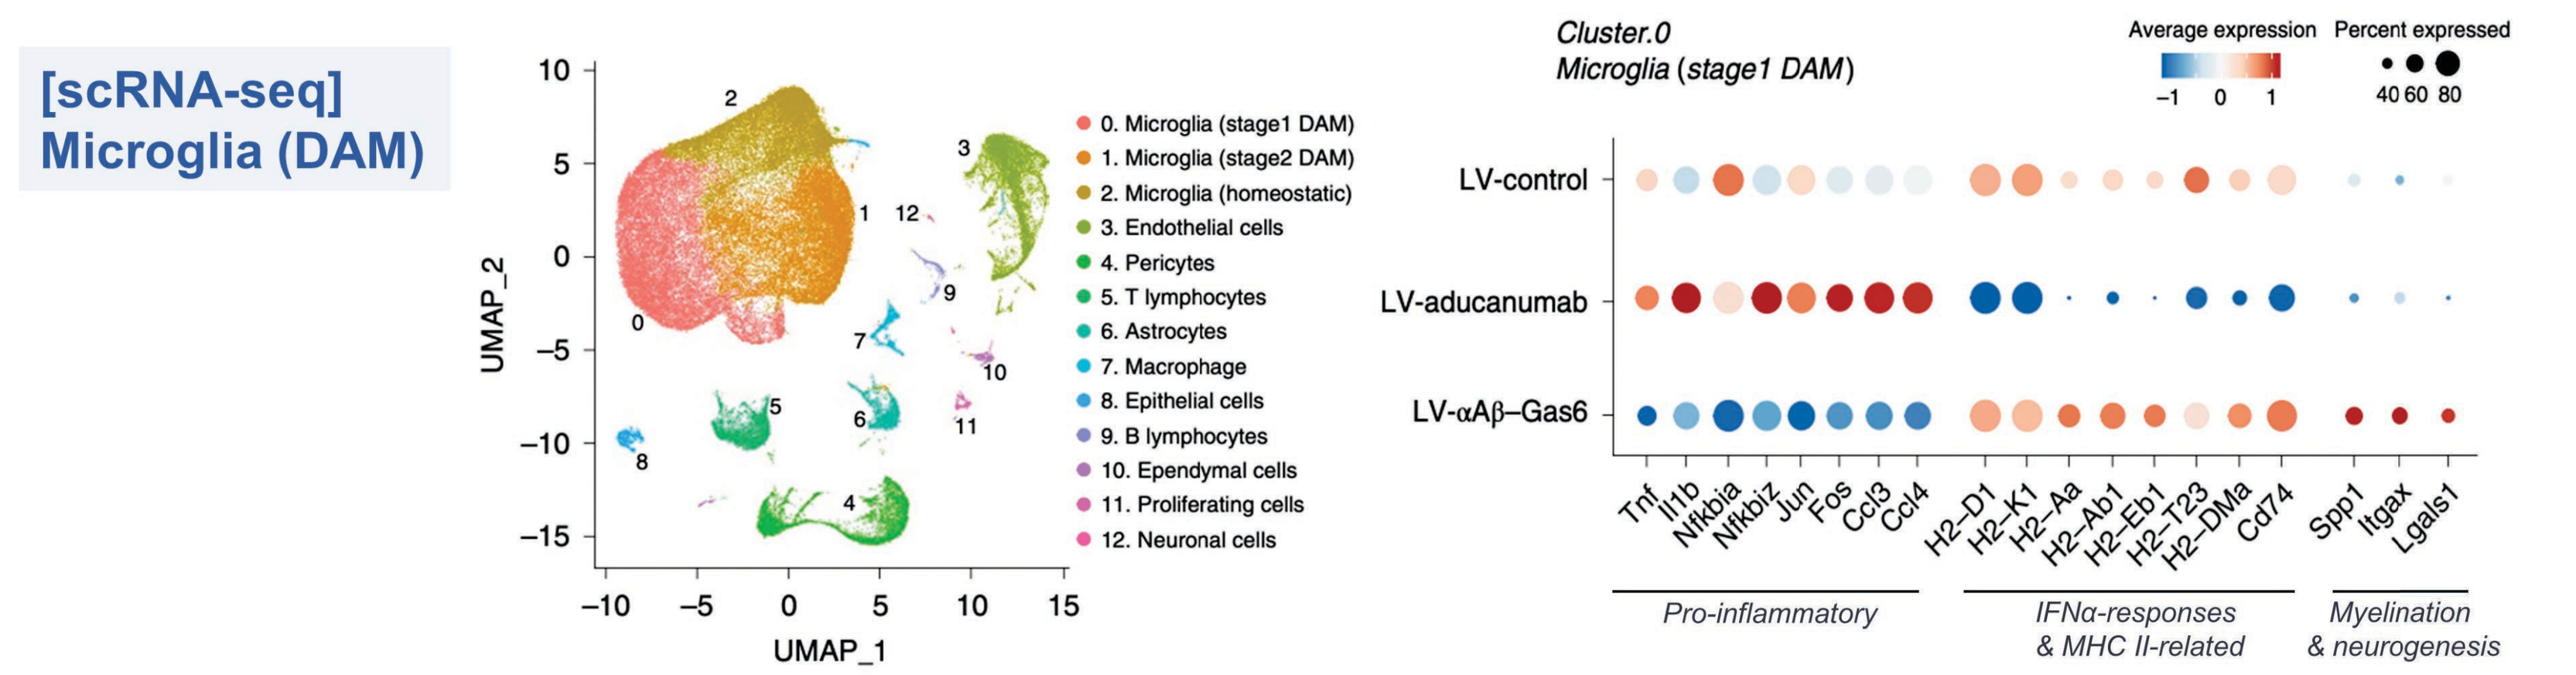
[scRNA-seq] Microglia (DAM)
UMAP_1
UMAP_2
0. Microglia (stage1 DAM)
1. Microglia (stage2 DAM)
2. Microglia (homeostatic)
3. Endothelial cells
4. Pericytes
5. T lymphocytes
6. Astrocytes
7. Macrophage
8. Epithelial cells
9. B lymphocytes
10. Ependymal cells
11. Proliferating cells
12. Neuronal cells
Cluster.0 Microglia (stage1 DAM)
Average expression
Percent expressed
LV-control
LV-aducanumab
LV-αAβ–Gas6
Tnf
Il1b
Nfkbia
Nfkbiz
Jun
Fos
Ccl3
Ccl4
H2–D1
H2–K1
H2–Aa
H2–Ab1
H2–Eb1
H2–T23
H2–DMa
Cd74
Spp1
Itgax
Lgals1
Pro-inflammatory
IFNα-responses & MHC II-related
Myelination & neurogenesis

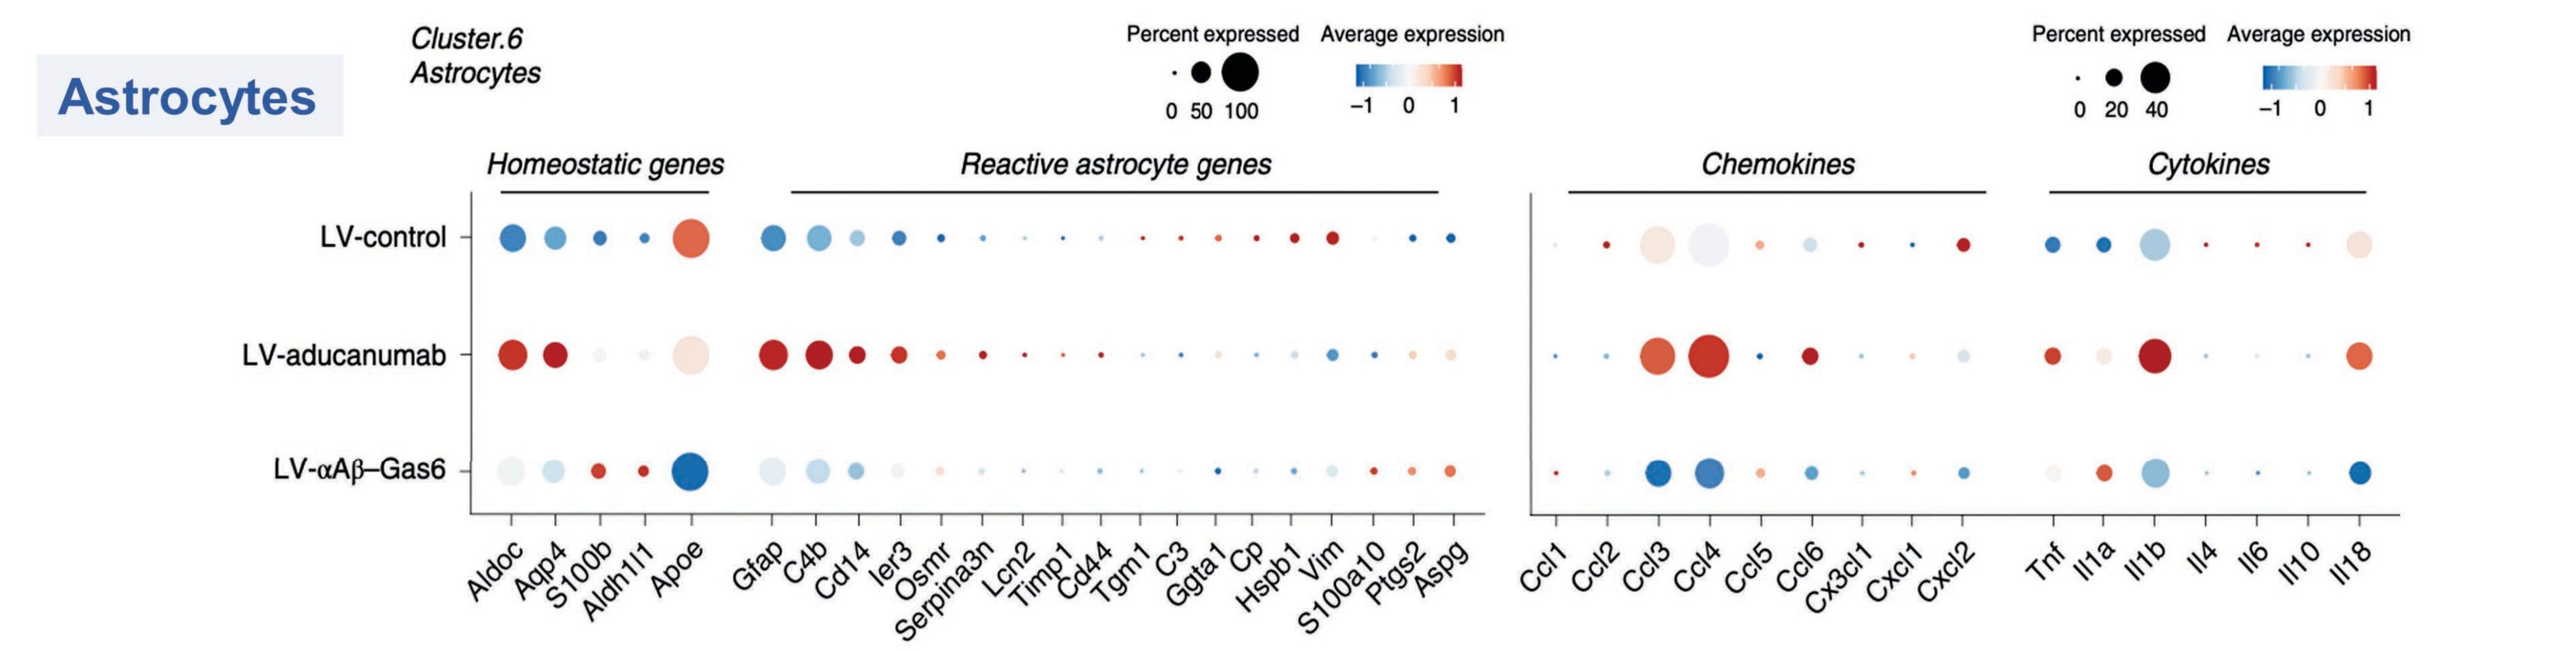
Astrocytes
Cluster.6 Astrocytes
Percent expressed
Average expression
Homeostatic genes
Reactive astrocyte genes
Chemokines
Cytokines
LV-control
LV-aducanumab
LV-αAβ–Gas6
Aldoc
Aqp4
S100b
Aldh1l1
Apoe
Gfap
C4b
Cd14
Ier3
Osmr
Serpina3n
Lcn2
Timp1
Cd44
Tgm1
C3
Ggta1
Cp
Hspb1
Vim
S100a10
Ptgs2
Aspg
Ccl1
Ccl2
Ccl3
Ccl4
Ccl5
Ccl6
Cx3cl1
Cxcl1
Cxcl2
Tnf
Il1a
Il1b
Il4
Il6
Il10
Il18

Microglia

- **Pro-inflammatory cytokines (Tnf and Il1b), chemokines (Ccl3, Ccl4, and Ccl6) as well as the molecules involved in NF-kB signaling (Nfkbia and Nfkbiz) and stress responses (Jun and Fos)** were all significantly upregulated in the LV-aducanumab group but were further suppressed in the LV-αAβ-Gas6-injected group.
- Microglia from the LV-αAβ-Gas6-injected group upregulated different gene sets related to **interferon-alpha responses and MHC class II-related genes (Cd74, H2-Aa, H2-D1, H2-k1, H2-Eb1, and so on)**. Furthermore, **neonatal microglial genes (Spp1, Itgax, Lgals1)** known to deliver signals for **myelination and neurogenesis** were also upregulated in the LV-αAβ–Gas6-injected group.

Astrocytes

- Similar to microglia, we found that astrocytes from the LV-aducanumab-injected group significantly upregulated **several reactive astrocyte genes (Gfap, C4b, and Cd14)** as well as **pro-inflammatory chemokines and cytokines** compared with the LV-αAβ-Gas6-injected group.

CONCLUSIONS

Harnessing the Power of TAM Biology

Improving cognitive function
- ✓ Preventing neuronal death & synaptic loss

More efficient Aβ clearance by utilizing astrocytes
- ✓ Expressing TAM receptor but no FcγR
- ✓ 2X larger number than microglia

Specific targeting of "inflammatory cells in inflammatory site"
- ✓ Only activated with target engaged
- ✓ Axl expression induced by inflammatory stimuli

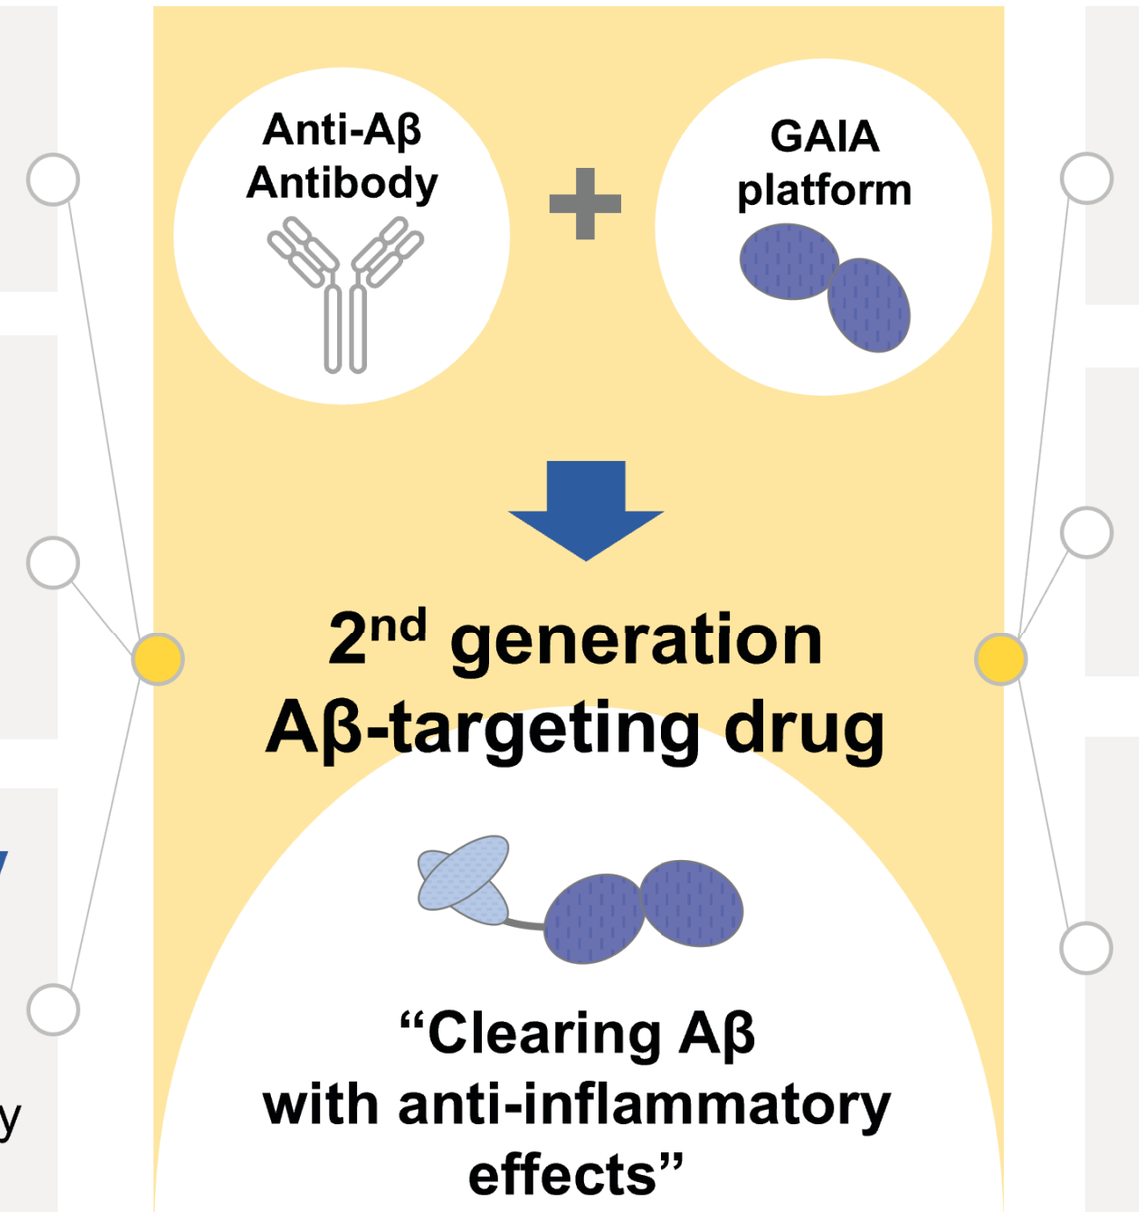
Anti-Aβ Antibody
GAIA platform
2nd generation Aβ-targeting drug
"Clearing Aβ with anti-inflammatory effects"

Reduced or no ARIA
- ✓ Glial modulation through TAM receptor significantly reduces ARIA

Broad therapeutic window
- ✓ Improved safety profile does not limit dose range anymore

Expanded market
- ✓ Safety profile of GAIA-Aβ suggests it can also treat high-risk patients
 1) with CAA or taking anti-coagulants
 2) APOE4 carrier
 3) showing ARIA after Mab treatment

*** The data presented here have been previously published in (Chung et al., Nature Medicine, 2022, PMID: 35927581). We are excited to share our results and encourage interested readers to refer to the publication for more detailed information.**

Created with BioRender Poster Builder